Point-of-care CART manufacture and delivery for the treatment of multiple myeloma and AL amyloidosis: the experience of Hadassah Medical Center

M. Assayag[1], N. Asherie[1], S. Kfir Erenfeld[1], B. Avni[1], T. Dubnikov[1], N. Zalcman[1], E. Lebel[2], E. Zimran[2], A. Shaulov[2], M. Pick[2], Y. Cohen[3], I. Avivi[3], C. Cohen[4], M. Gatt[2], S. Grisariu[1], P. Stepensky[1].

[1]Department of Bone Marrow Transplantation and Cancer Immunotherapy, Hadassah Medical Center, Faculty of Medicine, Hebrew University of Jerusalem, Jerusalem, Israel, [2]Department of Hematology, Hadassah Medical Center, Faculty of Medicine, Hebrew University of Jerusalem, Jerusalem, Israel, [3]Department of Hematology, Tel Aviv Medical Center, Sackler Faculty of Medicine, Tel Aviv University, Tel Aviv, Israel, [4]Laboratory of Tumor Immunology and Immunotherapy, The Mina and Everard Goodman Faculty of Life Sciences, Bar-Ilan University, Ramat Gan, Israel

INTRODUCTION

Anti-BCMA chimeric antigen receptor (CAR) T-cells for the treatment of multiple myeloma (MM) have proven safe and efficacious. Recently, two commercial products (i.e., ABECMA™ and CARVYKTI™) have been approved by the FDA and EMA agencies, auguring a rapid expansion of these "living-drugs" to the market. However, this is not the case yet, since access to this modality of treatment remains very limited. The lack of production slots, manufacturing costs and logistical issues are major drawbacks of this therapy and present significant obstacles to meeting the growing demand.

Point-of-care CART manufacture and delivery is an especially attractive approach for boosting the supply of CART products. In Israel, about 500 cases of myeloma are newly diagnosed every year, and about 100 cases will become candidates for CART therapy (after 3 previous lines). At Hadassah Medical Center (HMC) we have developed a new anti-BCMA CAR (HBI0101) for the treatment of MM and light chain amyloidosis (AL). CART products are manufactured and delivered to patients in-house, considerably shortening the "vein-to-vein" waiting time, which is critical especially for rapidly deteriorating patients.

RESULTS

The results of the phase Ia/b study (NCT04720313), exploring the safety and efficacy effects in 58 patients (50 MM- and 8 AL- patients) at escalating doses ranging from 150- to 800x10^6 CAR+ cells are presented here. For the MM cohort, at data cutoff of 09/02/2023, the median follow-up (mFU) for all doses was 174 days (range 18-531), with a median progression-free survival (mPFS) of 322 days and a median overall survival (mOS) not reached for the higher dose. The overall response rate for all cohorts was 82%, with a 87% response rate in the 800x10^6 cohort. Our results also suggest a trend toward better PFS and OS for non-exposed MM patients to prior BCMA-targeted therapies (PBTT). Amyloidosis patients with organ involvement and treated at low (N=1), intermediate (N=2) and high (N=5) doses, achieved complete responses including organ responses, demonstrating the manageable safety profile and efficacy of HBI0101 in the treatment of AL as well.

HBI0101 efficacy results in MM Patients

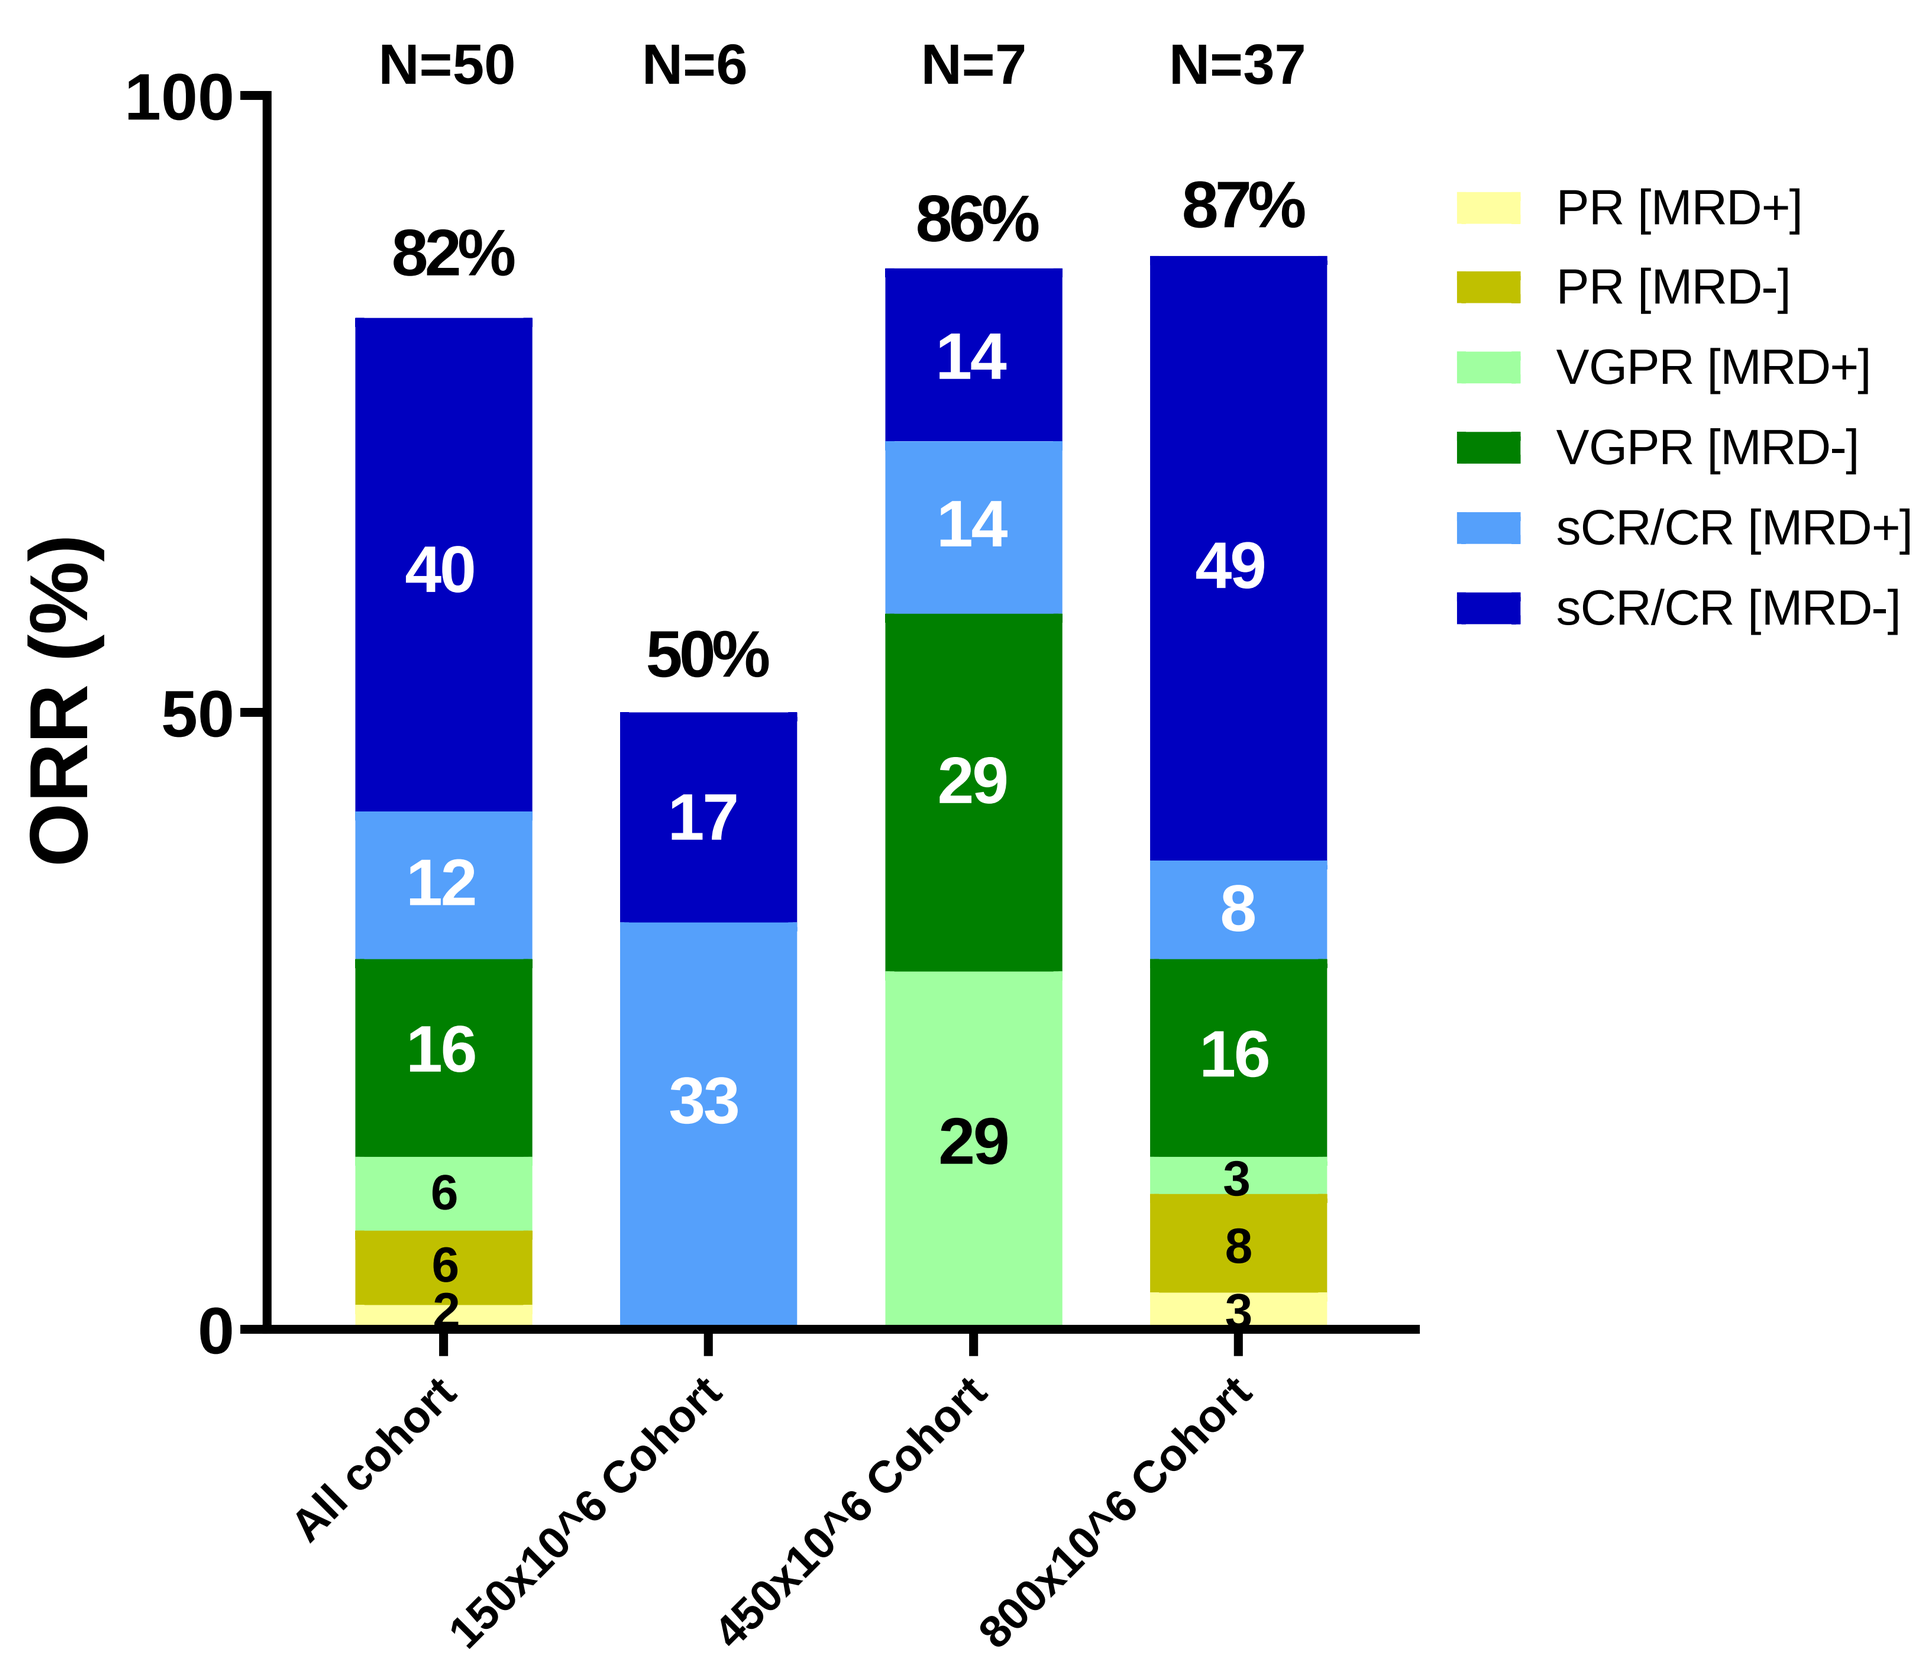
N=50
N=6
N=7
N=37
ORR (%)
82%
50%
86%
87%
All cohort
150x10^6 Cohort
450x10^6 Cohort
800x10^6 Cohort
PR [MRD+]
PR [MRD-]
VGPR [MRD+]
VGPR [MRD-]
sCR/CR [MRD+]
sCR/CR [MRD-]

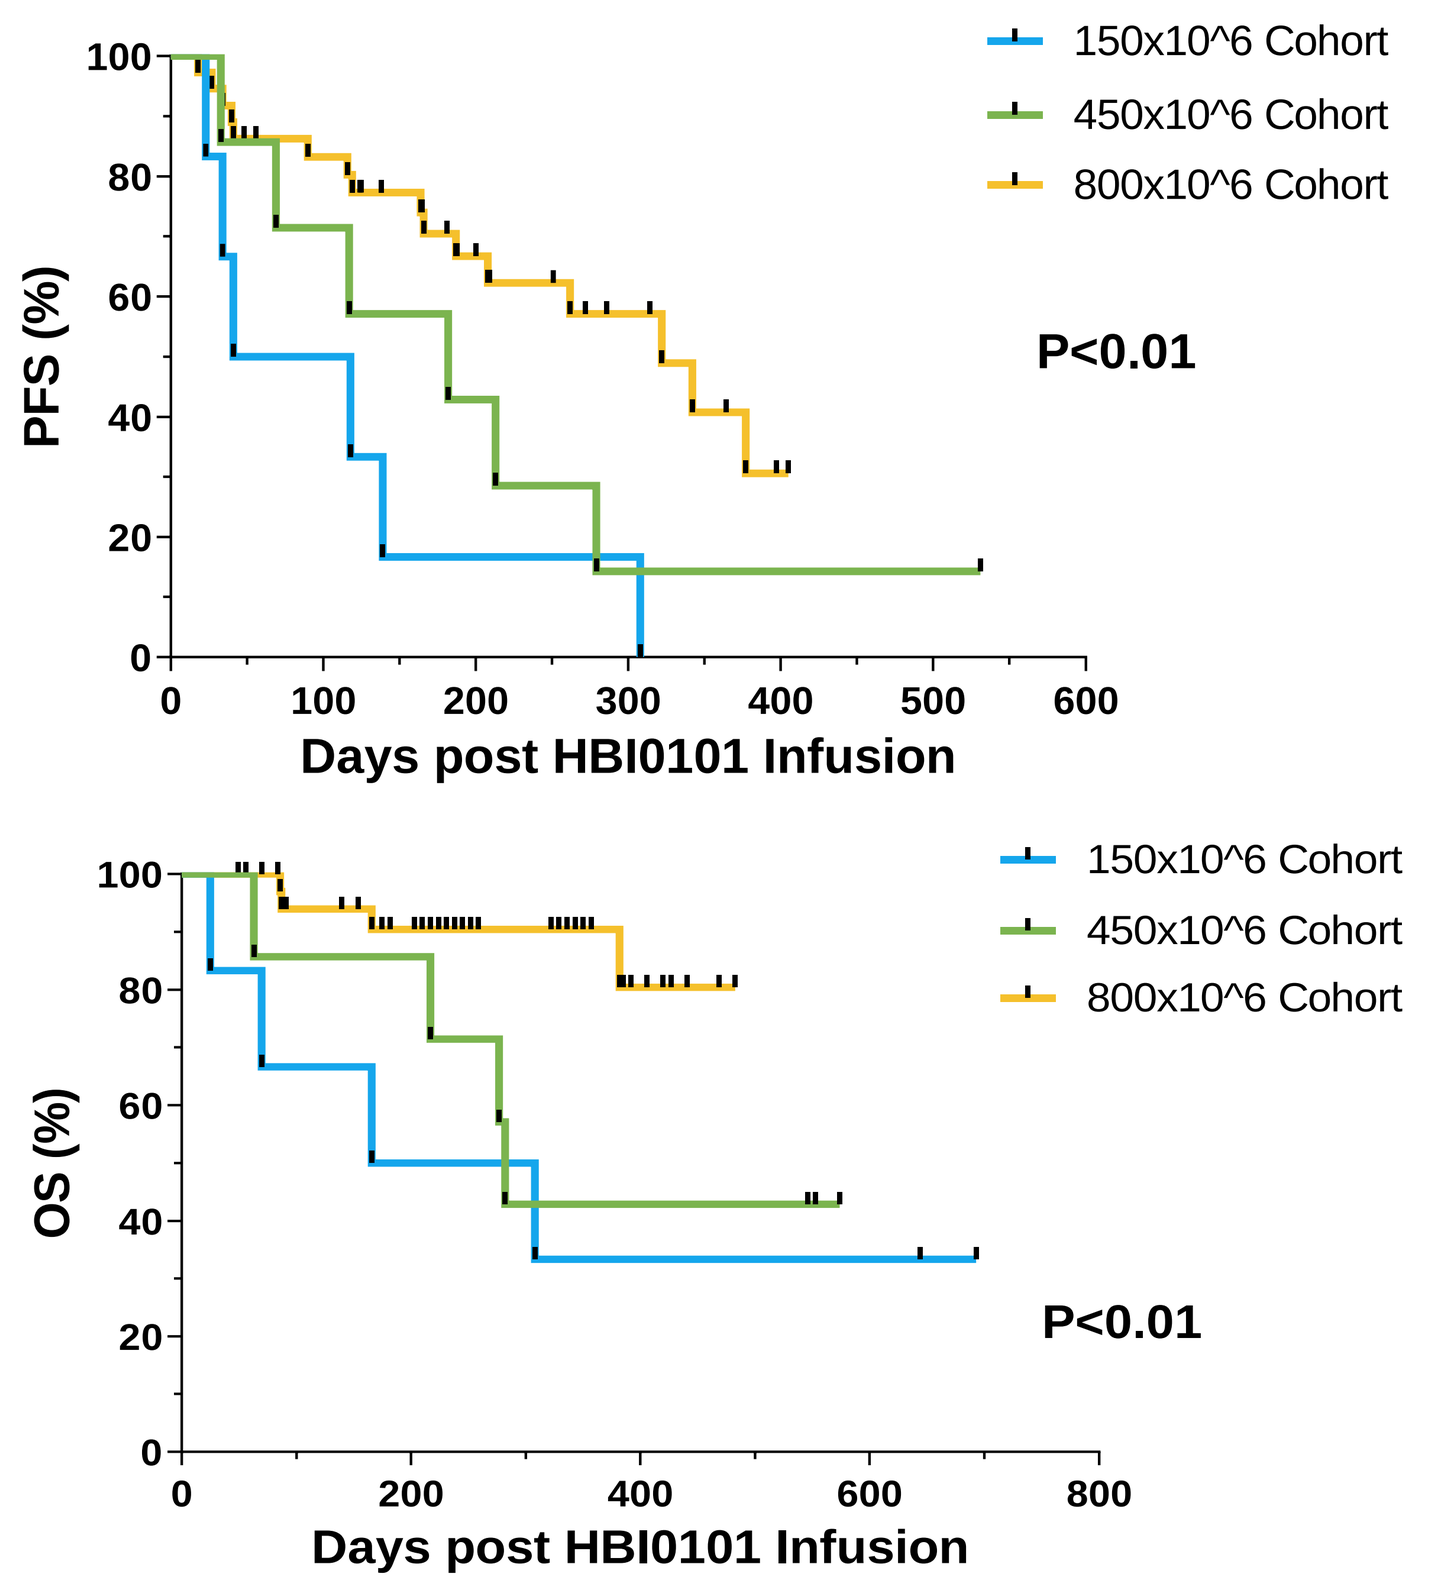
PFS (%)
OS (%)
150x10^6 Cohort
450x10^6 Cohort
800x10^6 Cohort
P<0.01
P<0.01
Days post HBI0101 Infusion
Days post HBI0101 Infusion

Figure 1: The percentage of PFS and OS are significantly improved in the 800x10^6 cohort compared to the low- and intermediate-dose cohorts.

	Median follow up (days)
150	80 (23-308)
450	182 (33-531)
800	187 (18-405)

Effect of BCMA-targeted prior therapy (PBTT) on MM patients' response to HBI0101 treatment (800x10^6 cohort)

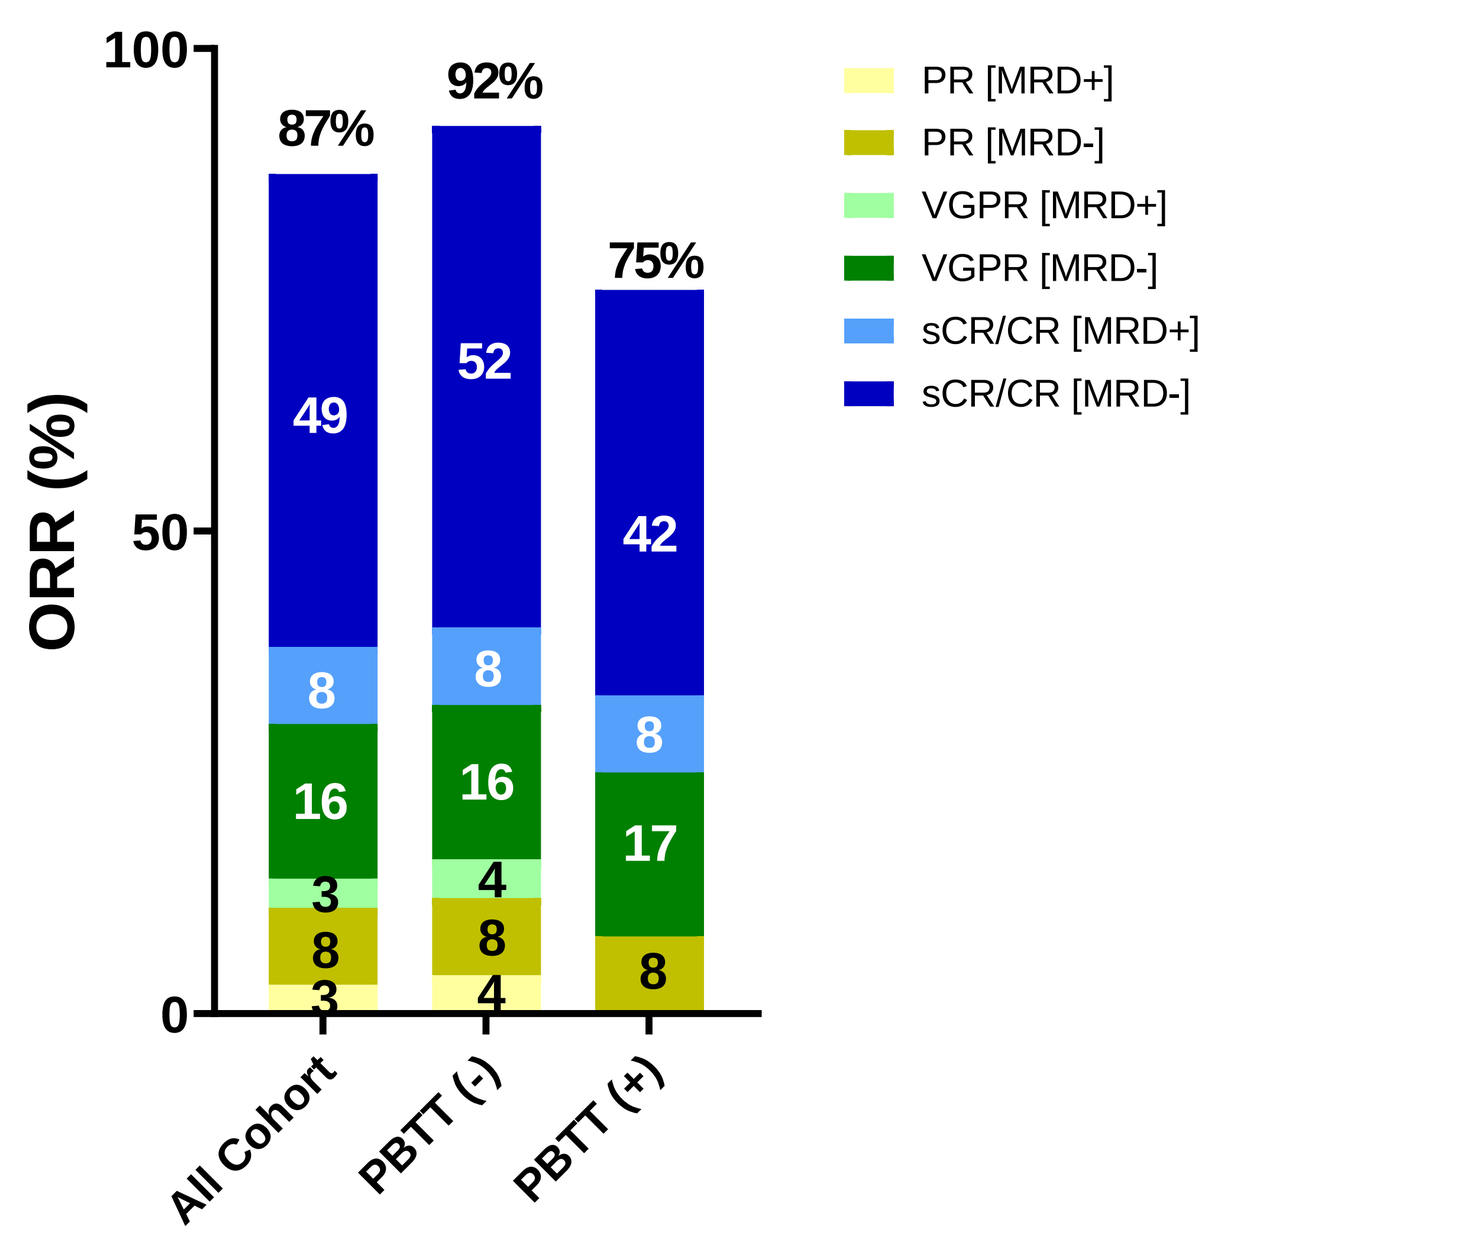
ORR (%)
87%
92%
75%
All Cohort
PBTT (-)
PBTT (+)
PR [MRD+]
PR [MRD-]
VGPR [MRD+]
VGPR [MRD-]
sCR/CR [MRD+]
sCR/CR [MRD-]

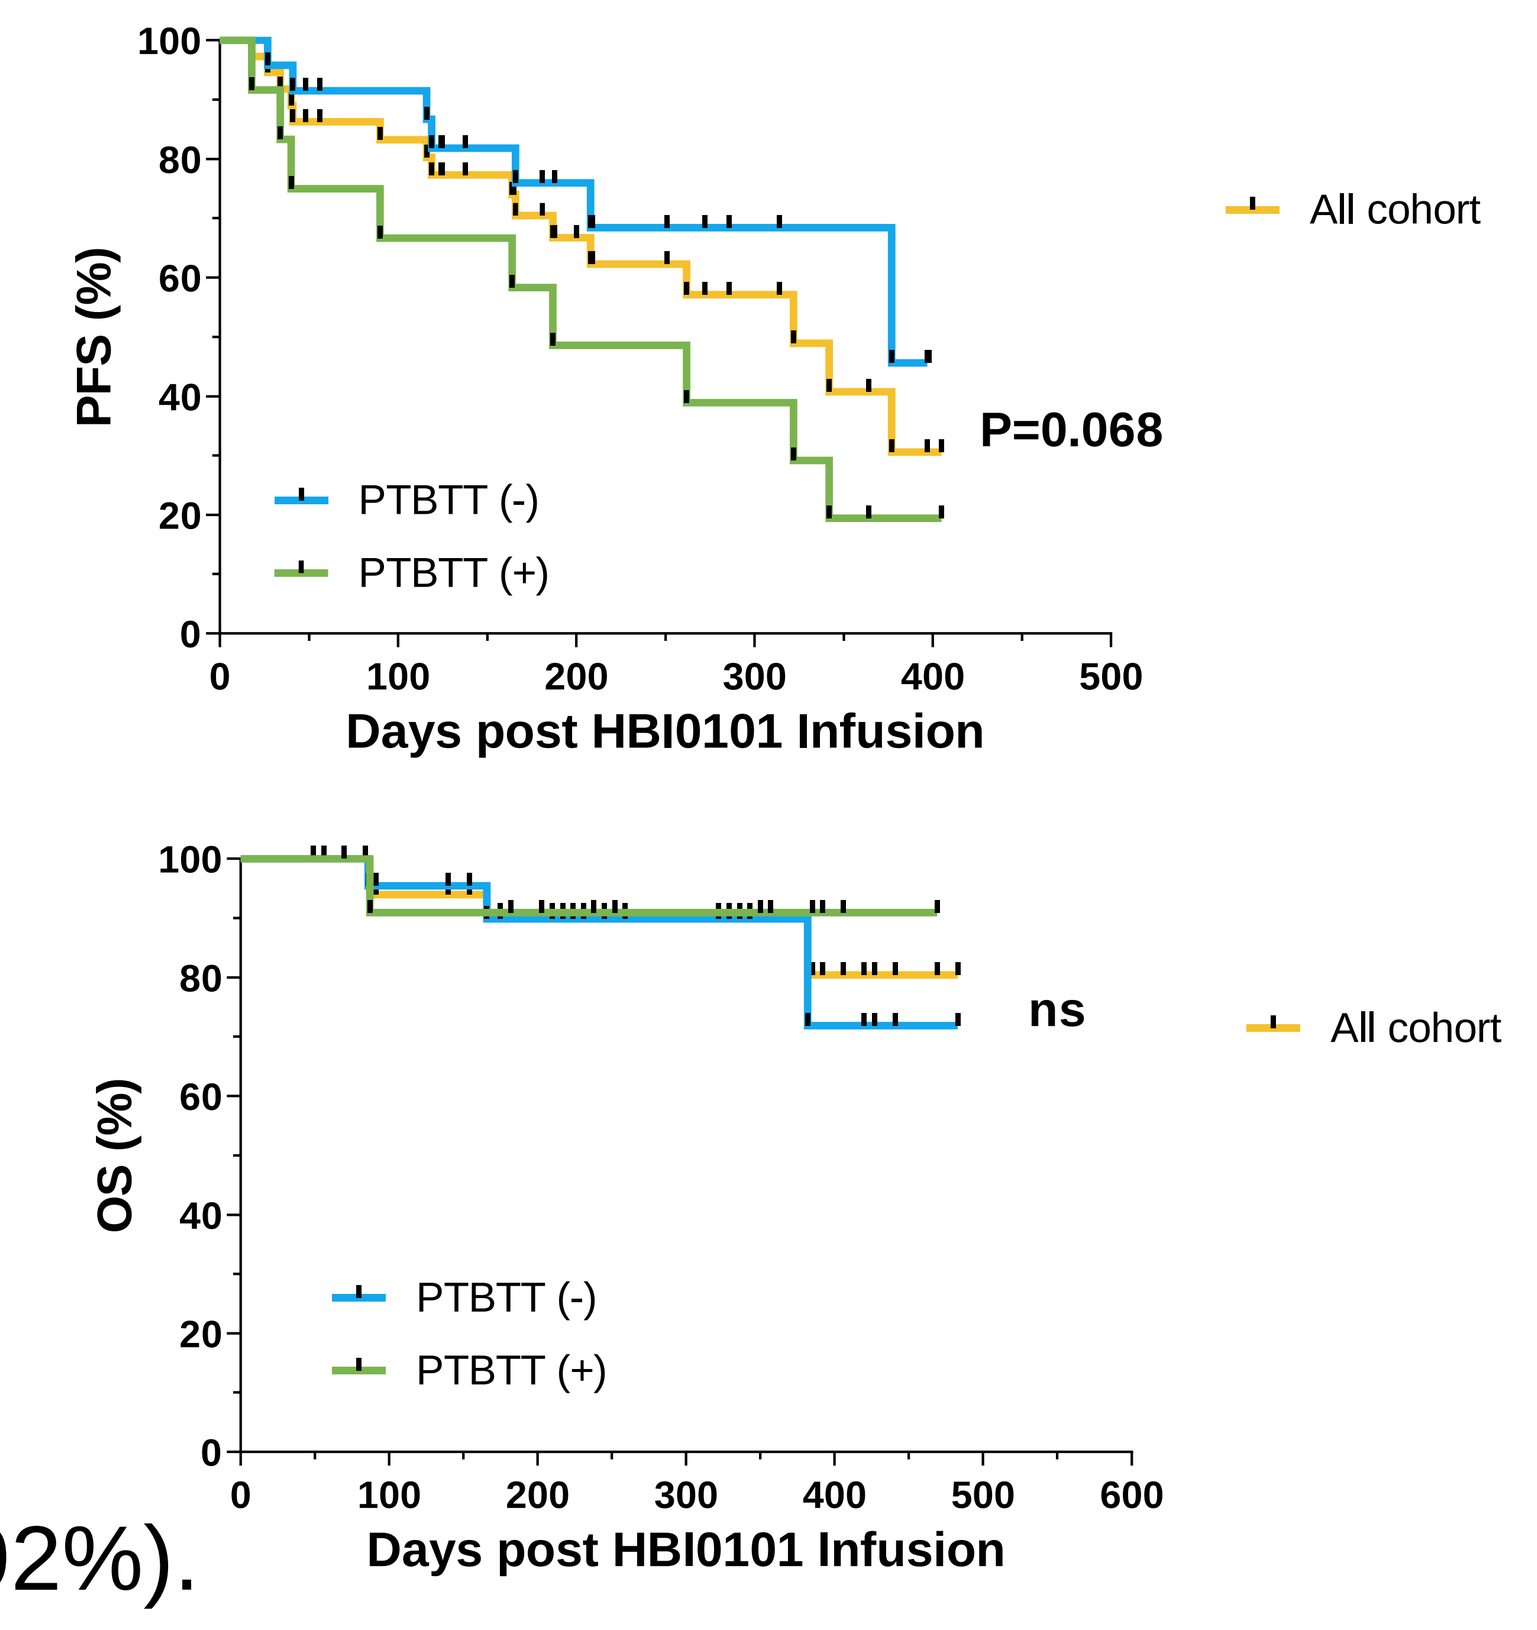
PFS (%)
OS (%)
All cohort
PTBTT (-)
PTBTT (+)
P=0.068
ns
Days post HBI0101 Infusion
Days post HBI0101 Infusion

Figure 2: Previous exposure to anti-BCMA therapy results in less favorable response rates (75% *vs.* 92%).

HBI0101 efficacy results in AL Patients

	Patient 1 *	Patient 2	Patient 3	Patient 4 **	Patient 5 *	Patient 6	Patient 7 **	Patient 8
Age	64	58	82	63	64	72	60	68
Gender	M	F	M	M	M	F	F	M
Organ involvement	Cardiac, Renal, Autonomic	Cardiac, Renal, Hepatic	Renal, GI	Cardiac, Hepatic, Lung, Soft tissue, Autonomic	Soft tissue, PNS	Cardiac, Renal, Liver	Cardiac, Soft Tissue	Cardiac, Renal, Soft Tissue
dFLC (mg/L)	**143**	**177**	**50**	**550**	**51**	**103**	**196**	**408**
ProBNP (pg/mL)	7500	2008	119	2773	731	28000	6600	220
Prior lines of therapy	8	6	6	10 and MDS	3	4	4	7
Years since diagnosis	10.5	4	15	4.5	2	3.5	0.8	11
CART+ infused (x10^6)	150	450	800	450	800	800	800	800
Organ Response	**Yes**	**Yes**	**Yes**	**Yes**	**No**	**No**	**Yes**	**NA**
dFLC (mg/L) at best response	**0**	**0**	**0**	**1.4**	**0.2**	**20**	**50**	**30**
MRD (10^{-5}) Negativity at Day 30 / Day 180	Yes / Yes	Yes / Yes	Yes / Yes	Yes	Yes	No	N/A	No

Figure 3: Enrolled AL patients presented with organ involvement, and were heavily pretreated with a median 6 LOT (range 3-10); 2 patients presented with concomitant MM (*) and 2 patients were treated on a compassionate basis (**). While HBI0101-related toxicity was clinically manageable in all cases, 5/8 patients achieved CR, 2/8 VGPR and 1/8 PR. A significant organ response with significant dFLC reduction were observed in all treated AL patients.

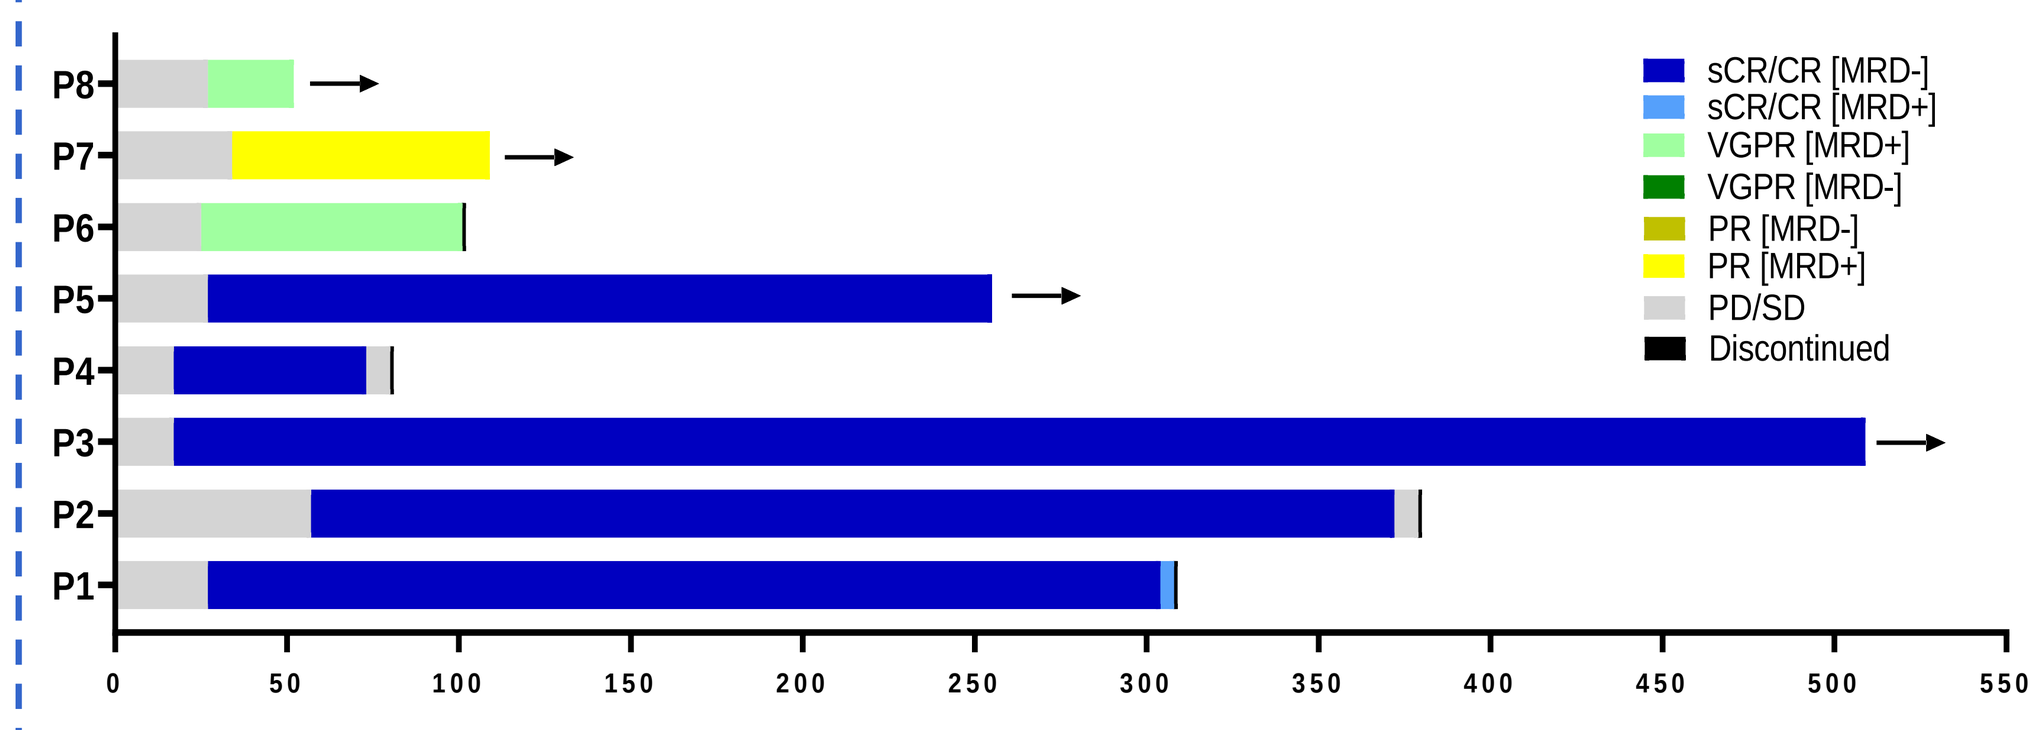
P8
P7
P6
P5
P4
P3
P2
P1
sCR/CR [MRD-]
sCR/CR [MRD+]
VGPR [MRD+]
VGPR [MRD-]
PR [MRD-]
PR [MRD+]
PD/SD
Discontinued

Swimmer's Plot analysis of AL Patients responses to HBI0101 (data cutoff: 02/04/2023). For the entire AL cohort, with a median FU period of 242 days (52-528), the median OS was 308 days (52-528).

HBI0101-related Toxicities MM and AL Patients

Figure 4: All patients displayed Grade 3-4 neutropenia and lymphopenia.

	Thrombocytopenia	Anemia	Neutropenia & Lymphopenia	Febrile Neutropenia	>28 d Hypoglobulinemia
MM COHORT					
Grade 1-2	8/50 (16%)	15/50 (30%)			33/50 (66%)
Grade 3-4	27/50 (54%)	30/50 (60%)	50/50 (100%)	34/50 (68%)	
AL COHORT					
Grade 1-2	1/8	1/8			8/8
Grade 3-4	1/8	1/8	8/8	3/8	

Cohort 1 – 150x10^6 6 Pt				Cohort 2 – 450x10^6 7 Pt				Cohort 3 – 800x10^6 37 Pt			
CRS 5/6 (83%)				CRS 6/7 (85%)				CRS 35/37 (94%)			
Grade 1		Grade 2		Grade 1		Grade 2		Grade 1	Grade 2	Grade 3	
4/6		1/6		2/7		4/7		10/37	20/37	5/37	
Tocilizumab Doses											
1	2	3	4	1	2	3	4	1	2	3	4
1/6	--	--	1/6	3/7	--	1/7	--	14/37	9/37	--	4/37
Hospitalization duration post infusion											
20.5 Days (14-166)				18 Days (14-25)				14 Days (10-27)			

CRS occurred in 92% (46/50) of MM patients and 75% (6/8) of AL patients, all clinically manageable. The vast majority of the CRS events started at the day of infusion.

The median duration of the CRS was 1 day (range 1-7) for MM the 800x10^6 dose and 3 days (1-5) for the 150 and 450x10^6 doses combined. Tocilizumab doses were required in 70% (33/46) of CRS events for MM patients. For AL patients, CRS was observed in 6/8 (75%) patients. No ICANS were recorded.

Slightly higher occurrence of CRS in the 800x106 dose treated patients, had no effect on the post infusion hospitalization duration.

CONCLUSION

HBI0101 CART product exhibits comparable safety and efficacy properties as the commercial anti-BCMA CART products reported in the literature (ABECMA™), with a clinically manageable safety profile and promising *in-vivo* efficacy. To the best of our knowledge, we present here the safety/efficacy results of an anti-BCMA CART based treatment in the largest AL cohort reported so far. Hadassah Medical Center's experience testifies of the feasibility of a decentralized approach for point-of-care CART manufacture to supply the local demand.

REFERENCES

1. *Harush O, Asherie N, Kfir-Erenfeld S, Adler G, Barliya T, Assayag M, Gatt ME, Stepensky P, Cohen CJ. Preclinical evaluation and structural optimization of anti-BCMA CAR to target multiple myeloma. Haematologica. 2022 Oct 1;107(10):2395-2407. doi: 10.3324/haematol.2021.280169. PMID: 35354252;*
2. *Kfir-Erenfeld S, Asherie N, Grisariu S, Avni B, Zimran E, Assayag M, Sharon TD, Pick M, Lebel E, Shaulov A, Cohen YC, Avivi I, Cohen CJ, Stepensky P, Gatt ME. Feasibility of a Novel Academic BCMA-CART (HBI0101) for the Treatment of Relapsed and Refractory AL Amyloidosis. Clin Cancer Res. 2022 Dec 1;28(23):5156-5166. doi: 10.1158/1078-0432.CCR-22-0637. PMID: 36107221.*
3. *Asherie N, Kfir-Erenfeld S, Avni B, Assayag M, Dubnikov T, Zalcman N, Lebel E, Zimran E, Shaulov A, Pick M, Cohen Y, Avivi I, Cohen C, Gatt ME, Grisariu S, Stepensky P. Development and manufacturing of novel locally produced anti-BCMA CART cells for the treatment of relapsed/refractory multiple myeloma: phase I clinical results. Haematologica. 2022 Oct 6. doi: 10.3324/haematol.2022.281628. Epub ahead of print. PMID: 36200421.*

www.ebmt.org